Early Results From the Phase 1b/2 EPCORE CLL-1 Trial: Subcutaneous Epcoritamab in Patients With Richter's Syndrome

Arnon P. Kater,[1] Jing Christine Ye,[2] Jose Sandoval-Sus,[3] Mar Bellido,[4] Jacob Haaber Christensen,[5] Anthony R. Mato,[6] Ann Janssens,[7] Toshihiko Oki,[8] Daniela Hoehn,[9] Marcia Rios,[9] Alexandra Kuznetsova,[10] Rebecca Valentin,[10] Herbert Eradat[11]

[1]Amsterdam UMC, Cancer Center Amsterdam, University of Amsterdam, Amsterdam, Netherlands; [2]Rogel Cancer Center, University of Michigan, Ann Arbor, MI, USA; [3]Moffitt Cancer Center at Memorial Healthcare System, Pembroke Pines, FL, USA; [4]University Medical Center Groningen and University of Groningen, Groningen, Netherlands; [5]Odense University Hospital, Odense, Denmark; [6]Memorial Sloan Kettering Cancer Center, Chronic Lymphocytic Leukemia Program, New York, NY, USA; [7]University Hospitals Leuven, Leuven, Belgium; [8]AbbVie, North Chicago, IL, USA; [9]Genmab, Princeton, NJ, USA; [10]Genmab, Copenhagen, Denmark; [11]David Geffen School of Medicine at UCLA, Los Angeles, CA, USA

OBJECTIVE

To evaluate the preliminary clinical efficacy and safety of epcoritamab in patients with RS-DLBCL

CONCLUSIONS

In a difficult-to-treat, high-risk RS-DLBCL patient population, single-agent epcoritamab showed promising activity (high response rates: ORR 60%; CMR 50%) with a tolerable safety profile (only low-grade CRS, all resolved; no ICANS events; no discontinuations due to TEAEs)

This first standalone dataset for a bispecific antibody in RS supports further clinical exploration of epcoritamab in this aggressive disease with limited treatment options

The EPCORE CLL-1 study is ongoing and recruiting patients with CLL or RS (NCT04623541)

Copies of this poster obtained through quick response (QR) code are for personal use only and may not be reproduced without permission from the authors of this poster.

References

1. Wang Y, et al. *Haematologica*. 2020;105:765-73. **2.** Rossi D, et al. *Blood*. 2012;119:521-9. **3.** Langerbeins P, et al. *Am J Hematol*. 2014;89:E239-43. **4.** Rogers KA, et al. *Br J Haematol.* 2018;180:259-66. **5.** Younes A, et al. *Lancet Haematol*. 2019;6:e67-78. **6.** Engelberts PJ, et al. *EBioMedicine*. 2020;52:102625. **7.** van der Horst HJ, et al. *Blood Cancer J*. 2021;11:38. **8.** Hutchings M, et al. *Lancet*. 2021;398:1157-69. **9.** Thieblemont C, et al. *J Clin Oncol*. 2022;DOI:10.1200/JCO.22.01725. **10.** Kater AP, et al. ASH 2021. Abstract 2627.

Acknowledgments

On behalf of all the authors, we thank the patients, study investigators, and site personnel for their participation in this study. This study was funded by Genmab A/S and AbbVie. Medical writing and graphical support were provided by Peloton Advantage, LLC, an OPEN Health company, Parsippany, NJ, USA, and funded by Genmab. These results were previously presented at the American Society of Hematology Annual Meeting; December 10–13, 2022; New Orleans, LA, and virtual.

Disclosures

Kater: AbbVie: Honoraria, Advisory Board, Research Funding; Janssen, AstraZeneca: Advisory Board, Steering Committee, Research Funding; BMS, Roche/Genentech: Advisory Board, Research Funding; Genmab, Lava: Advisory Board, Steering Committee. **Ye:** BMS: Consultancy; Ascentage: Research Funding; Sanofi: Research Funding; Karyopharm: Research Funding; Portola: Research Funding; MingSight: Research Funding; Nektar: Research Funding; Janssen: Consultancy, Research Funding; GSK: Research Funding; Regeneron: Research Funding; Genmab: Research Funding. **Sandoval-Sus:** Seagen, Incyte, Janssen, ADC Therapeutics, Genentech, Genmab: Advisory Board; Massive Bio: Consultancy. **Bellido:** Amgen: Financial Contribution to Attend Workshop; Janssen: Financial Departmental Contribution for Educational Purposes. **Christensen:** There are no relationships to disclose. **Mato:** AbbVie: Consultancy, Research Funding; Adaptive Biotechnologies: Consultancy, Research Funding; Janssen: Consultancy, Research Funding; Pharmacyclics, an AbbVie Company: Consultancy, Research Funding; MSKCC: Current Employment; Genmab: Research Funding; Johnson & Johnson: Consultancy, Research Funding; Genentech: Consultancy, Research Funding; Loxo: Consultancy, Research Funding; Nurix: Research Funding; DTRM Biopharma: Consultancy, Research Funding; TG Therapeutics: Consultancy, DSMB, Research Funding; BeiGene: Consultancy, Research Funding; Acerta/AstraZeneca: Consultancy, Research Funding; AstraZeneca: Consultancy; Sunesis: Consultancy, Research Funding. **Janssens:** Genmab: Current Employment; AbbVie: Consultancy, Travel Grants, Speakers Bureau; Amgen: Consultancy, Travel Grants, Speakers Bureau; AstraZeneca: Consultancy, Speakers Bureau; BeiGene: Consultancy, Speakers Bureau; Janssen: Consultancy, Speakers Bureau; Incyte: Consultancy, Speakers Bureau; Novartis: Consultancy, Speakers Bureau; Sobi: Consultancy, Speakers Bureau; Sanofi: Consultancy, Speakers Bureau; Roche: Consultancy; Celgene: Travel Grants. **Oki:** AbbVie: Current Employment. **Hoehn:** Genmab: Current Employment. **Rios:** Genmab: Current Employment. **Kuznetsova:** Genmab: Current Employment. **Valentin:** Genmab: Current Employment. **Eradat:** Genentech: Consultancy, Honoraria, Research Funding, Speakers Bureau; AbbVie: Consultancy, Honoraria, Research Funding, Speakers Bureau; Incyte: Consultancy, Honoraria, Research Funding, Speakers Bureau; MorphoSys: Consultancy, Honoraria, Research Funding, Speakers Bureau; Pharmacyclics: Honoraria, Research Funding; Juno: Research Funding; BeiGene: Consultancy, Honoraria, Research Funding, Speakers Bureau; Gilead: Research Funding; Kite: Research Funding; AstraZeneca: Research Funding; Celgene: Research Funding; BMS: Research Funding; Atara: Research Funding.

BACKGROUND

- Richter's syndrome (RS) is the development of an aggressive lymphoma from underlying chronic lymphocytic leukemia (CLL) or small lymphocytic lymphoma (SLL)
 - Occurs in approximately 2–10% of patients with CLL/SLL,[1] and most commonly, disease transforms to $CD20^+$ diffuse large B-cell lymphoma (DLBCL)
- RS-DLBCL is characterized by sudden clinical deterioration
 - Poor prognostic factors include prior treatment for CLL/SLL, elevated lactate dehydrogenase, and adverse molecular abnormalities or cytogenetics (eg, *TP53, NOTCH1*, trisomy 12, and del17p)[1,2]
 - Chemoimmunotherapies have limited responses[3,4]
 - Median overall survival ranges from a few months to approximately 1 year[1,4,5]
 - There is no established standard of care; current guidelines recommend clinical trials
- High unmet medical need exists for novel, efficacious, and well-tolerated therapies
- Epcoritamab (DuoBody®-CD3xCD20) is a subcutaneous bispecific antibody that binds to CD3 on T cells and CD20 on B cells to induce T-cell–mediated killing of $CD20^+$ malignant B cells[6,7]
- Single-agent epcoritamab in large B-cell lymphoma (LBCL) in the EPCORE NHL-1 pivotal trial:
 - Epcoritamab demonstrated substantial antitumor efficacy with a well-tolerated safety profile in relapsed or refractory, including heavily pretreated, patients with LBCL[8,9]; trial included one patient with RS-DLBCL
- Herein, we present initial data from the RS cohort in the ongoing phase 1b/2 EPCORE CLL-1 trial, the first standalone dataset for a bispecific antibody in Richter's syndrome

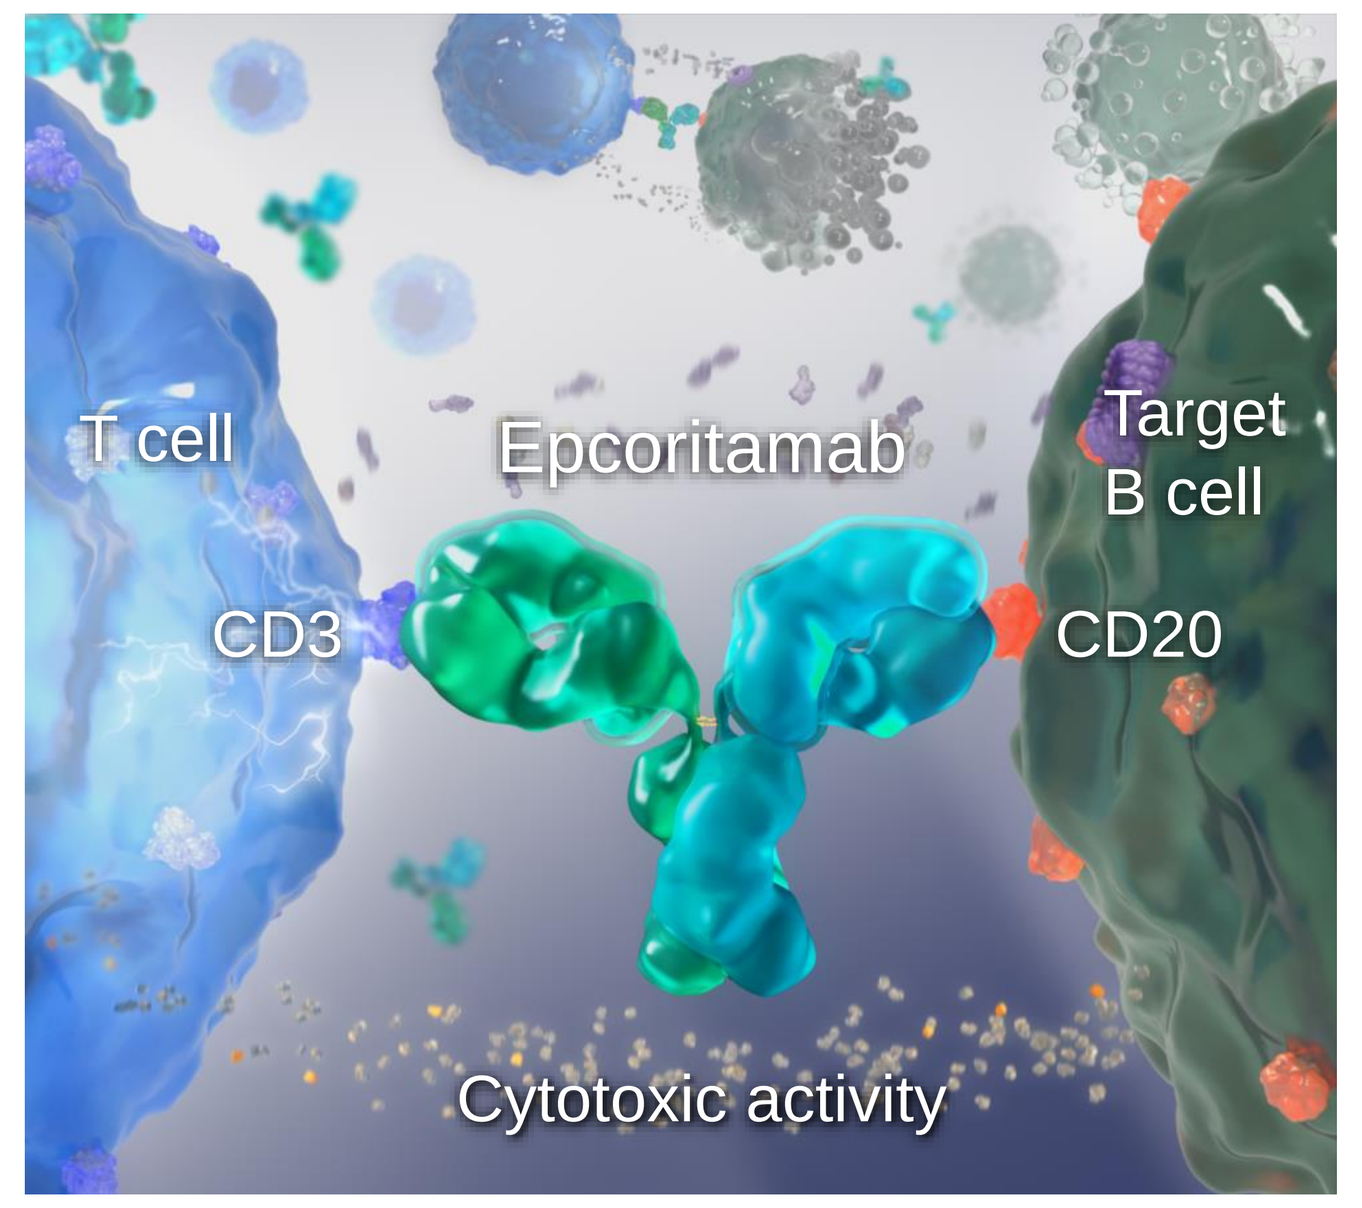
T cell
Epcoritamab
Target B cell
CD3
CD20
Cytotoxic activity

STUDY DESIGN: EPCORE CLL-1 RS Expansion Cohort

Dose escalation

Median follow-up: 4.9 mo (range, 0.6–9.3)

CLL[10]

- ✓ No DLTs
- ✓ MTD not reached
- ✓ RP2D identified
- ✓ Manageable safety profile
- ✓ Encouraging antitumor activity

Key RS inclusion criteria

- Ineligible for or voluntarily declined chemotherapy
- ≤1 prior line of therapy for RS-DLBCL
- Biopsy-proven transformation to $CD20^+$ RS-DLBCL
- Prior clinical history of CLL or SLL
- ECOG PS 0–2
- Measurable disease by PET and/or CT/MRI

RS-DLBCL expansion, N=10

Step-up dosing[a] | **Epcoritamab (SC) 48 mg** QW C1–3, Q2W C4–9, Q4W C10+ → Efficacy assessment by PET-CT obtained Q6W until C6, and then Q24W thereafter → Treatment until disease progression

- To ensure patient safety and better characterize CRS, inpatient monitoring was required at first 4 doses of epcoritamab
- **Primary endpoint:** Overall response rate (ORR)
- **Key secondary endpoints:** Complete metabolic response (CMR) rate, time to response (TTR), and safety/tolerability

Data cutoffs: September 8, 2022 (efficacy); September 16, 2022 (safety). Epcoritamab was administered in 28-d cycles as shown. [a]Patients received SC epcoritamab with step-up dosing (ie, 0.16 mg priming and 0.8 mg intermediate doses before first full dose) and corticosteroid prophylaxis as previously described to mitigate CRS.

RESULTS

Patient Characteristics and Treatment History

CLL Characteristic, n (%)	Total, N=10
IGHV unmutated[a]	8 (80)
TP53 mutation[b]	5 (50)
NOTCH1 mutation[c]	2 (20)
FISH	
Trisomy 12[d]	1 (10)
Del17p[e]	3 (30)
Del11q[f]	3 (30)
Del13q[g]	4 (40)
RS Characteristic	**Total, N=10**
Median age, y (range)	70 (53–79)
Male, n (%)	7 (70)
Ann Arbor stage, n (%)	
IE	1 (10)
II	1 (10)
III	3 (30)
IV	5 (50)
Elevated lactate dehydrogenase, n (%)	8 (80)
Cell of origin, n (%)[h]	
Germinal center B-cell	1 (10)
Non–germinal center/Activated B-cell	6 (60)
Characteristic of Prior CLL Therapy	**Total, N=10**
Median time from initial CLL diagnosis to first dose, y (range)	12 (2.6–24.0)
Median number of prior lines of therapy for CLL (range)	**3 (0–7)**
Prior CLL/SLL therapy, n (%)	7 (70)
Chemoimmunotherapies	7 (70)
Targeted agents	6 (60)
BCL2 inhibitor	5 (50)
BTK inhibitor	5 (50)
CAR T-cell therapy	1 (10)
Median time from last CLL treatment to first dose, mo (range)	12 (0.2–61.8)
Characteristic of Prior RS-DLBCL Therapy	**Total, N=10**
1 prior line of RS-DLBCL therapy, n (%)	**5 (50)**
R-CHOP	3 (30)
No response	2 (20)
R-DHAP	1 (10)
No response	1 (10)
VR-EPOCH[i]	1 (10)
Median time from disease transformation to first dose, mo (range)	3.4 (0.5–21.4)
Median time from end of last line of RS-DLBCL therapy to first dose, mo (range)	2 (0.5–5.4)

Data cutoff: September 16, 2022. Data for CLL characteristics were obtained from local laboratories. [a]*IGHV* mutation status unknown for 2 patients. [b]*TP53* mutation status unmutated for 4 patients and unknown for 1 patient. [c]*NOTCH1* mutation status unmutated for 4 patients and unknown for 4 patients. [d]Trisomy 12 status negative for 8 patients and unknown for 1 patient. [e]Del17p status negative for 7 patients. [f]Del11q status negative for 7 patients. [g]Del13q status negative for 4 patients and unknown for 2 patients. [h]Cell of origin was unknown for 3 patients. [i]Response to VR-EPOCH unknown. R-CHOP, rituximab, cyclophosphamide, doxorubicin, vincristine, and prednisone; R-DHAP, rituximab, dexamethasone, cytarabine, and cisplatin; VR-EPOCH, venetoclax plus dose-adjusted rituximab, etoposide, prednisone, vincristine, cyclophosphamide, and doxorubicin.

- Prior novel therapies for CLL indicate a modern, difficult-to-treat, high-risk RS-DLBCL patient population; 1 patient received prior CAR T-cell therapy

Treatment-Emergent AEs (≥30%)

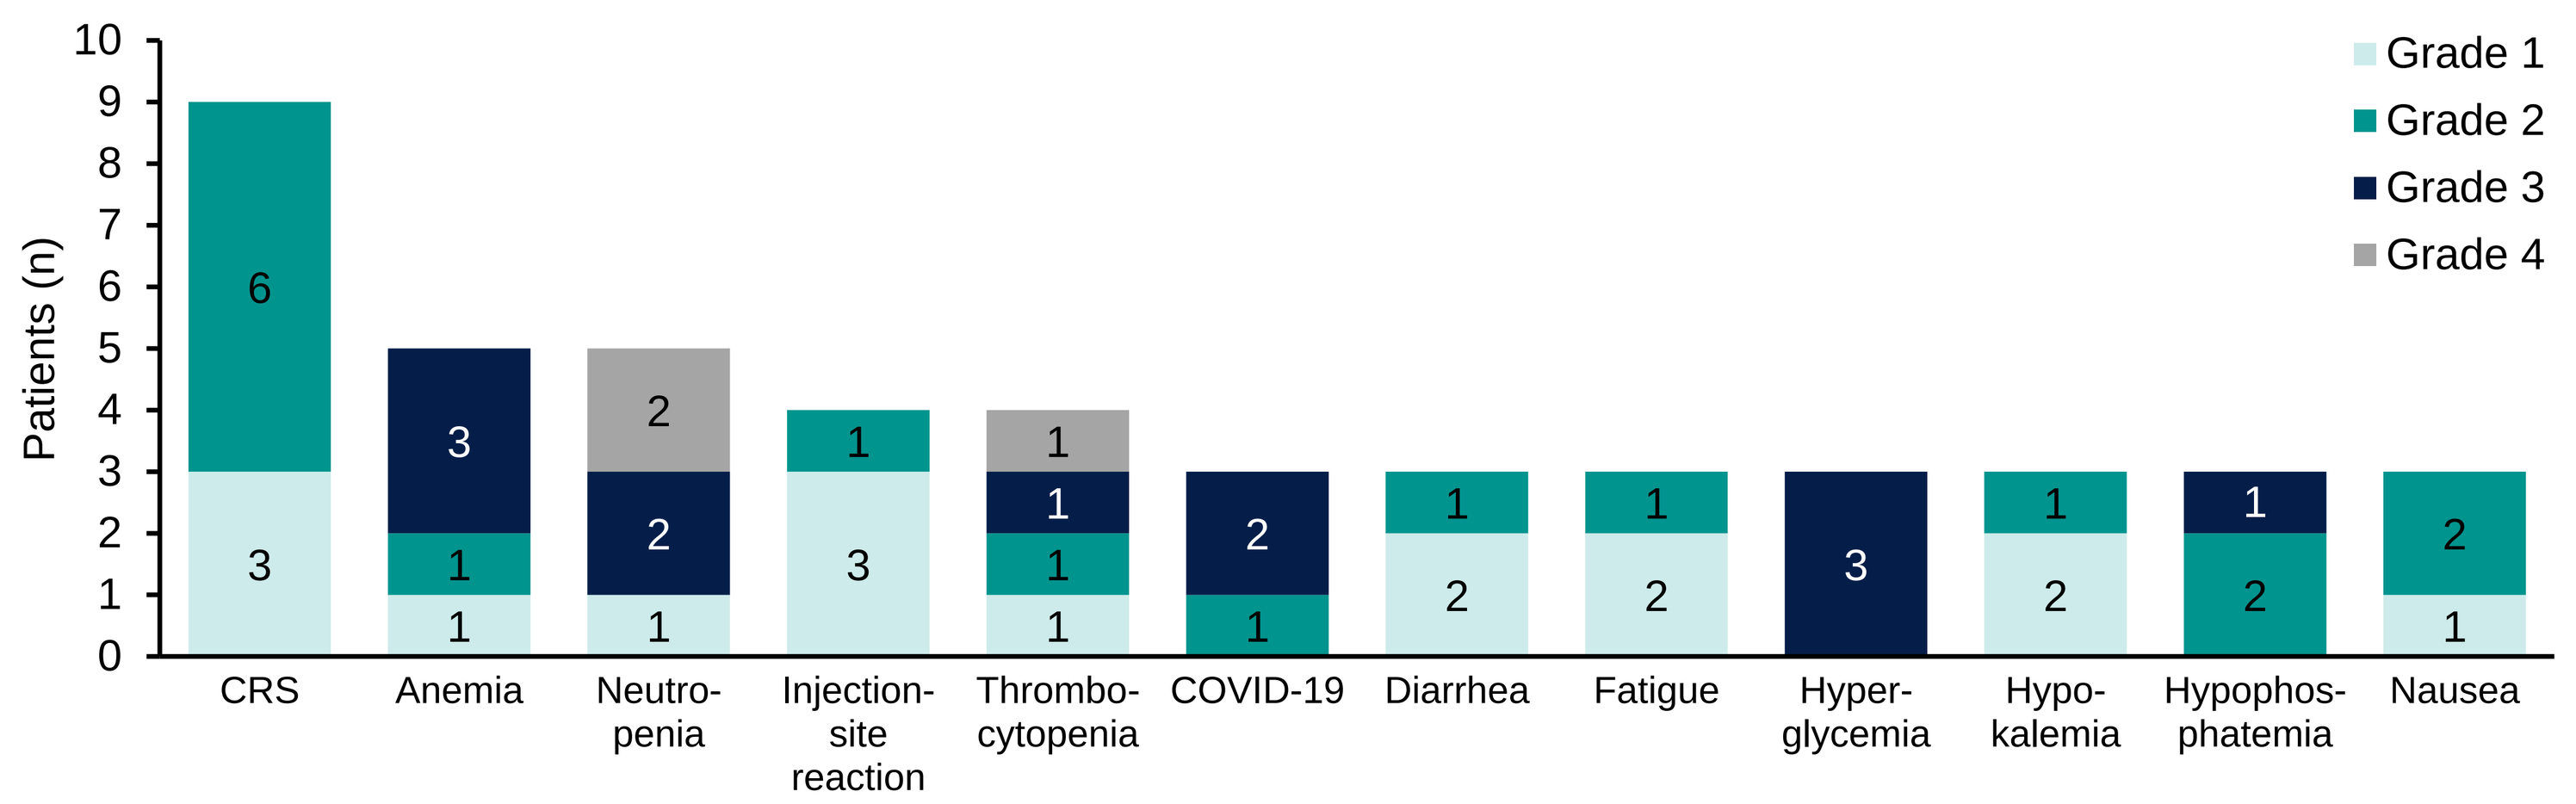
Patients (n)
Grade 1
Grade 2
Grade 3
Grade 4
CRS
Anemia
Neutro-penia
Injection-site reaction
Thrombo-cytopenia
COVID-19
Diarrhea
Fatigue
Hyper-glycemia
Hypo-kalemia
Hypophos-phatemia
Nausea

Data cutoff: September 16, 2022. Median duration of treatment: 3.5 mo (range, 0.5–9.3). Median cycles (28 d) of epcoritamab: 4 (range, 1–11).

- AEs were primarily low grade, including CRS; no ICANS events were observed
- One case of clinical tumor lysis syndrome (grade 2); resolved in 3 days
- One grade 5 (fatal) TEAE (general physical health deterioration in the setting of PD, unrelated to epcoritamab)
- Six patients experienced dose delay due to TEAE
- No TEAEs leading to discontinuation

CRS Events

	Total, N=10
CRS, n (%)[a]	9 (90)
Grade 1	3 (30)
Grade 2	6 (60)
CRS resolution, n/n (%)	**9/9 (100)**
Median time to onset after first full dose, h (range)	12.5 (8–394)
Median time to resolution, d (range)[b]	3 (2–9)
Treated with tocilizumab, n (%)	7 (70)
Leading to treatment discontinuation, n (%)	**0**

Data cutoff: September 16, 2022. [a]Graded by Lee et al 2019 criteria. [b]Median is Kaplan–Meier estimate based on longest CRS duration in patients with CRS.

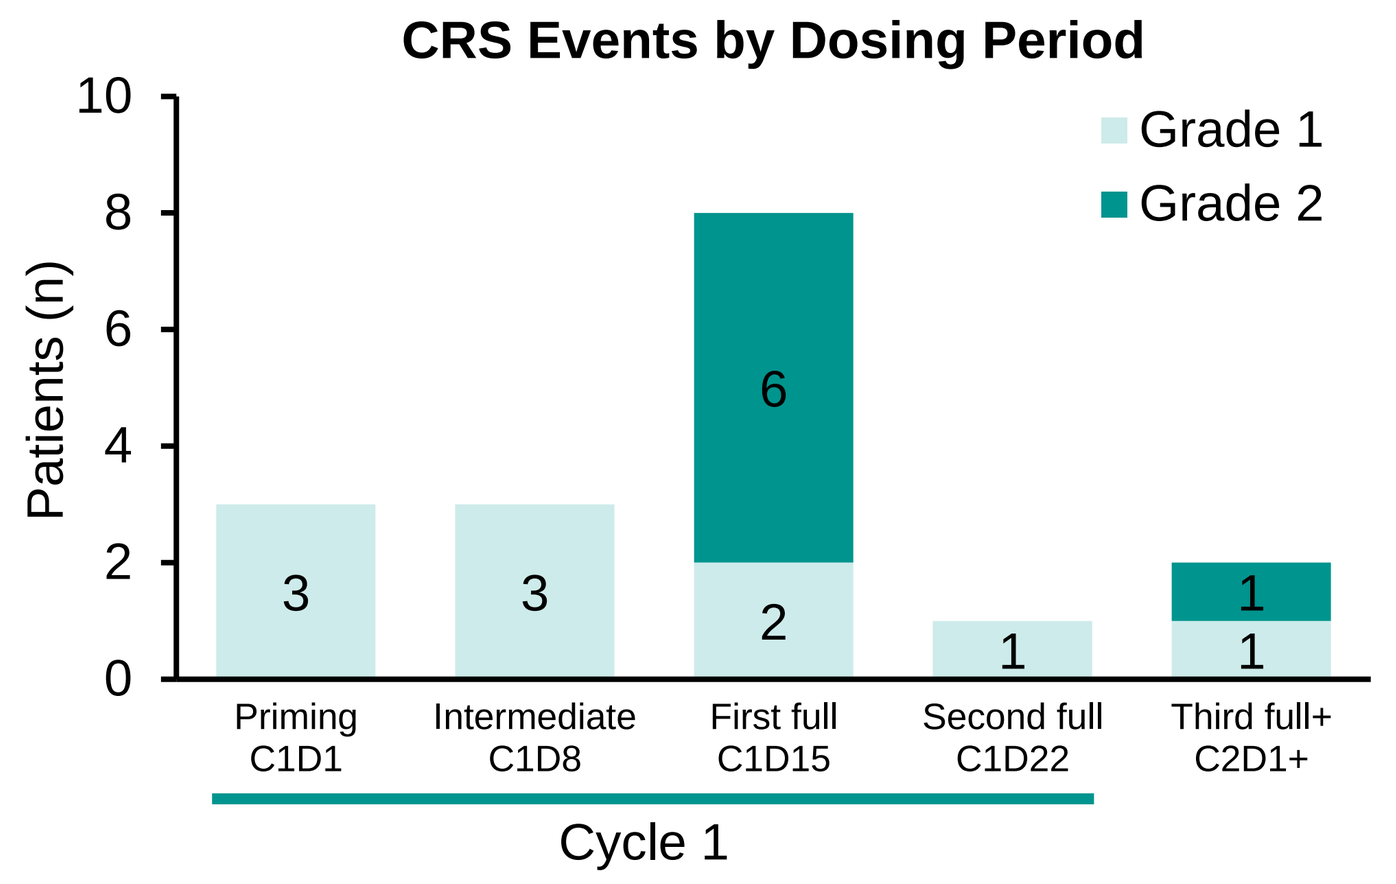
CRS Events by Dosing Period
Grade 1
Grade 2
Patients (n)
Priming C1D1
Intermediate C1D8
First full C1D15
Second full C1D22
Third full+ C2D1+
Cycle 1

Data cutoff: September 16, 2022.

- Occurrence was predictable, with most cases following the first full dose
- No grade ≥3 CRS events
- All CRS events resolved, and none led to discontinuation

Best Overall Response

Response, n (%)[a]	Total Efficacy Evaluable, N=10
Overall response[b]	**6 (60)**
Complete metabolic response (CMR)	5 (50)
Partial metabolic response (PMR)	1 (10)
Stable disease	1 (10)
Progressive disease	2 (20)
Not evaluable	1 (10)[c]

Data cutoff: September 8, 2022. Median follow-up: 4.9 mo (range, 0.6–9.3).
[a]Based on modified response-evaluable population, defined as patients with ≥1 target lesion at baseline and ≥1 postbaseline response evaluation and/or patients who died within 60 d of first dose.
[b]Response assessment according to Lugano 2014 criteria. [c]Patient stopped treatment on C1D15 due to progression and did not receive any scans.

- High overall and complete metabolic response rates observed

Best Tumor Reduction From Baseline

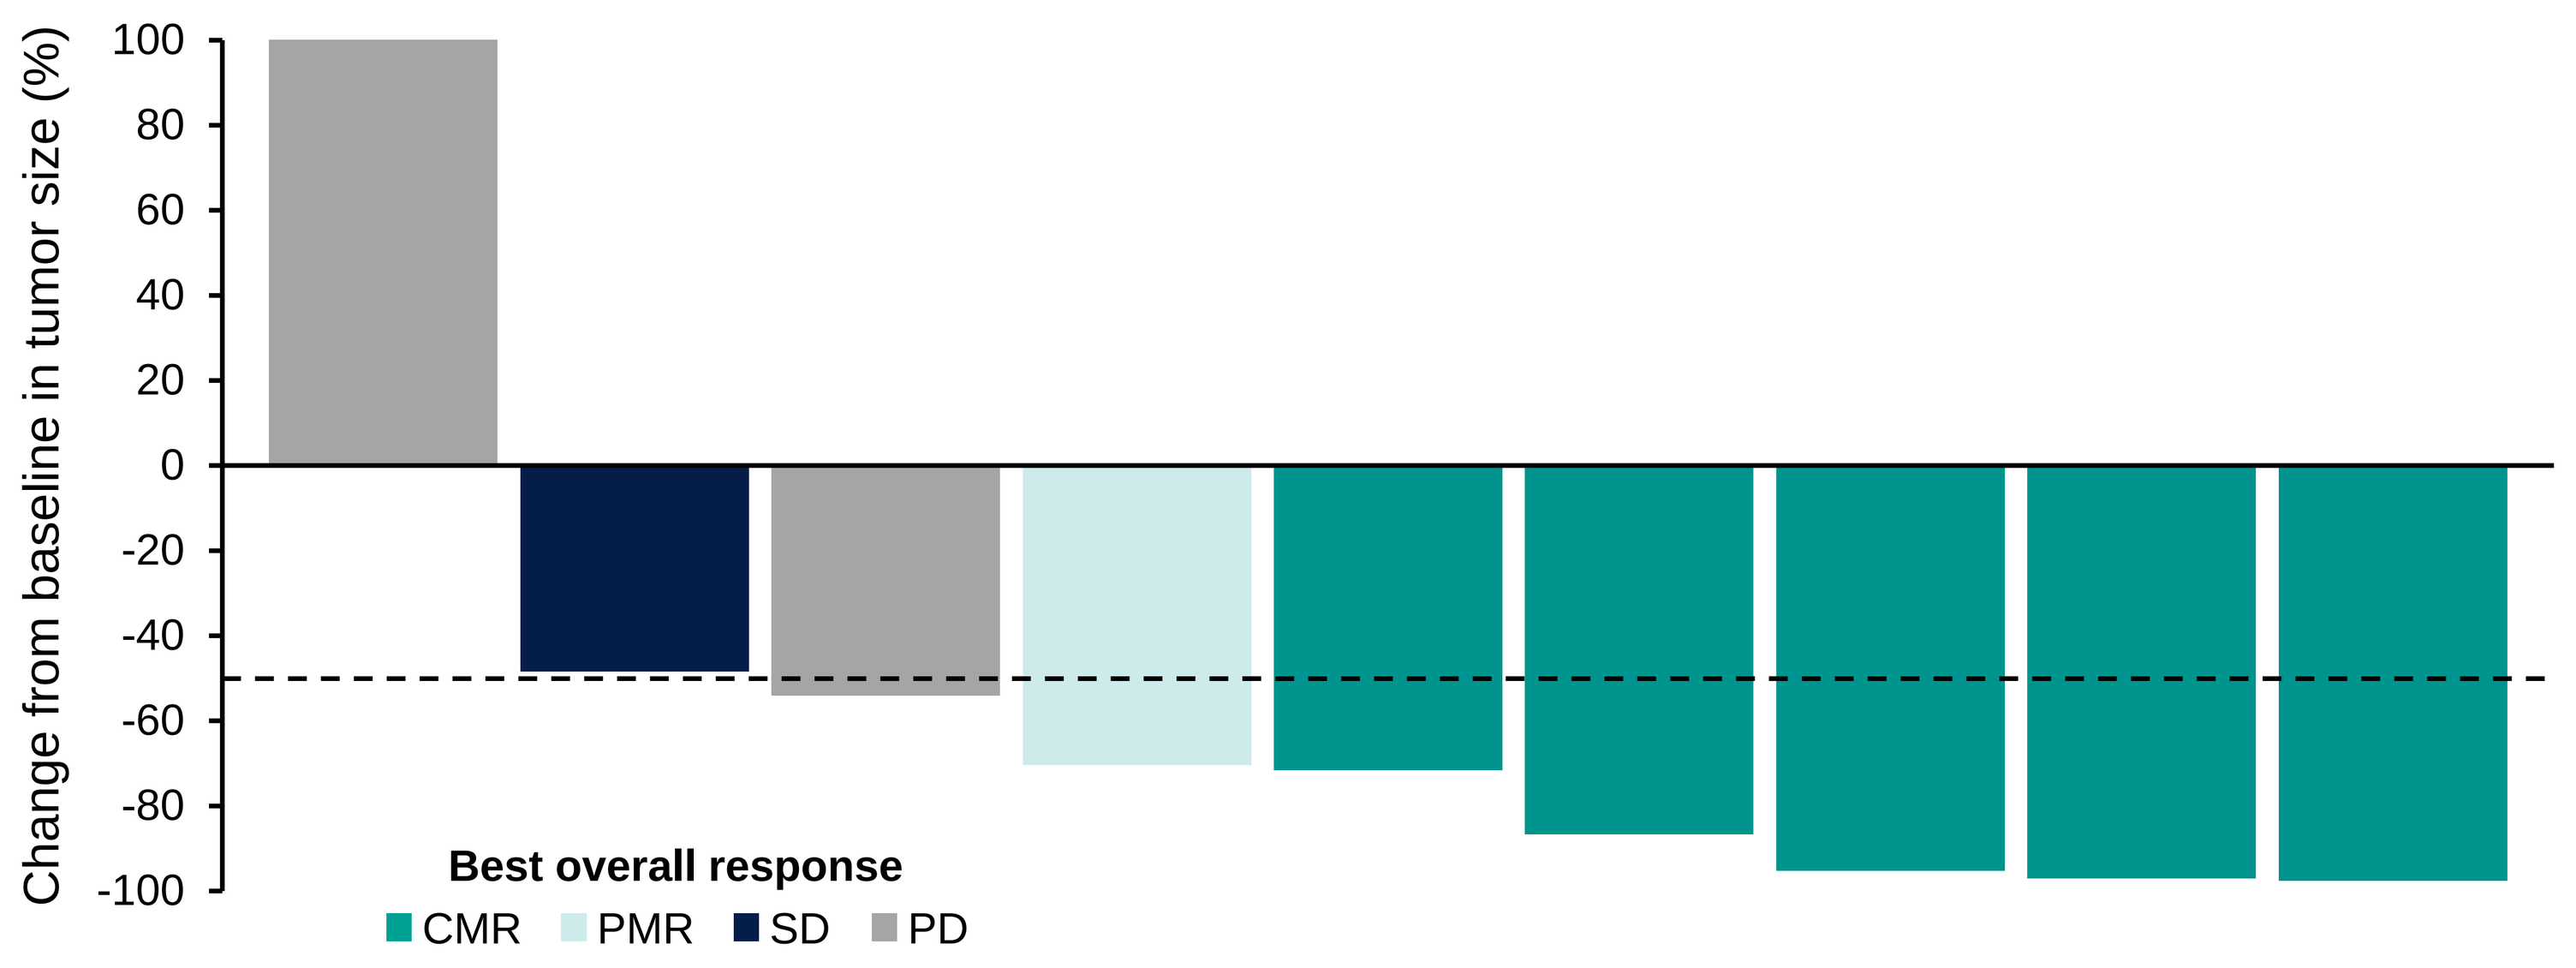
Change from baseline in tumor size (%)
Best overall response
CMR
PMR
SD
PD

Data cutoff: September 8, 2022.

- Nearly all patients achieved at least 50% reduction in tumor size from baseline

Depth and Duration of Response

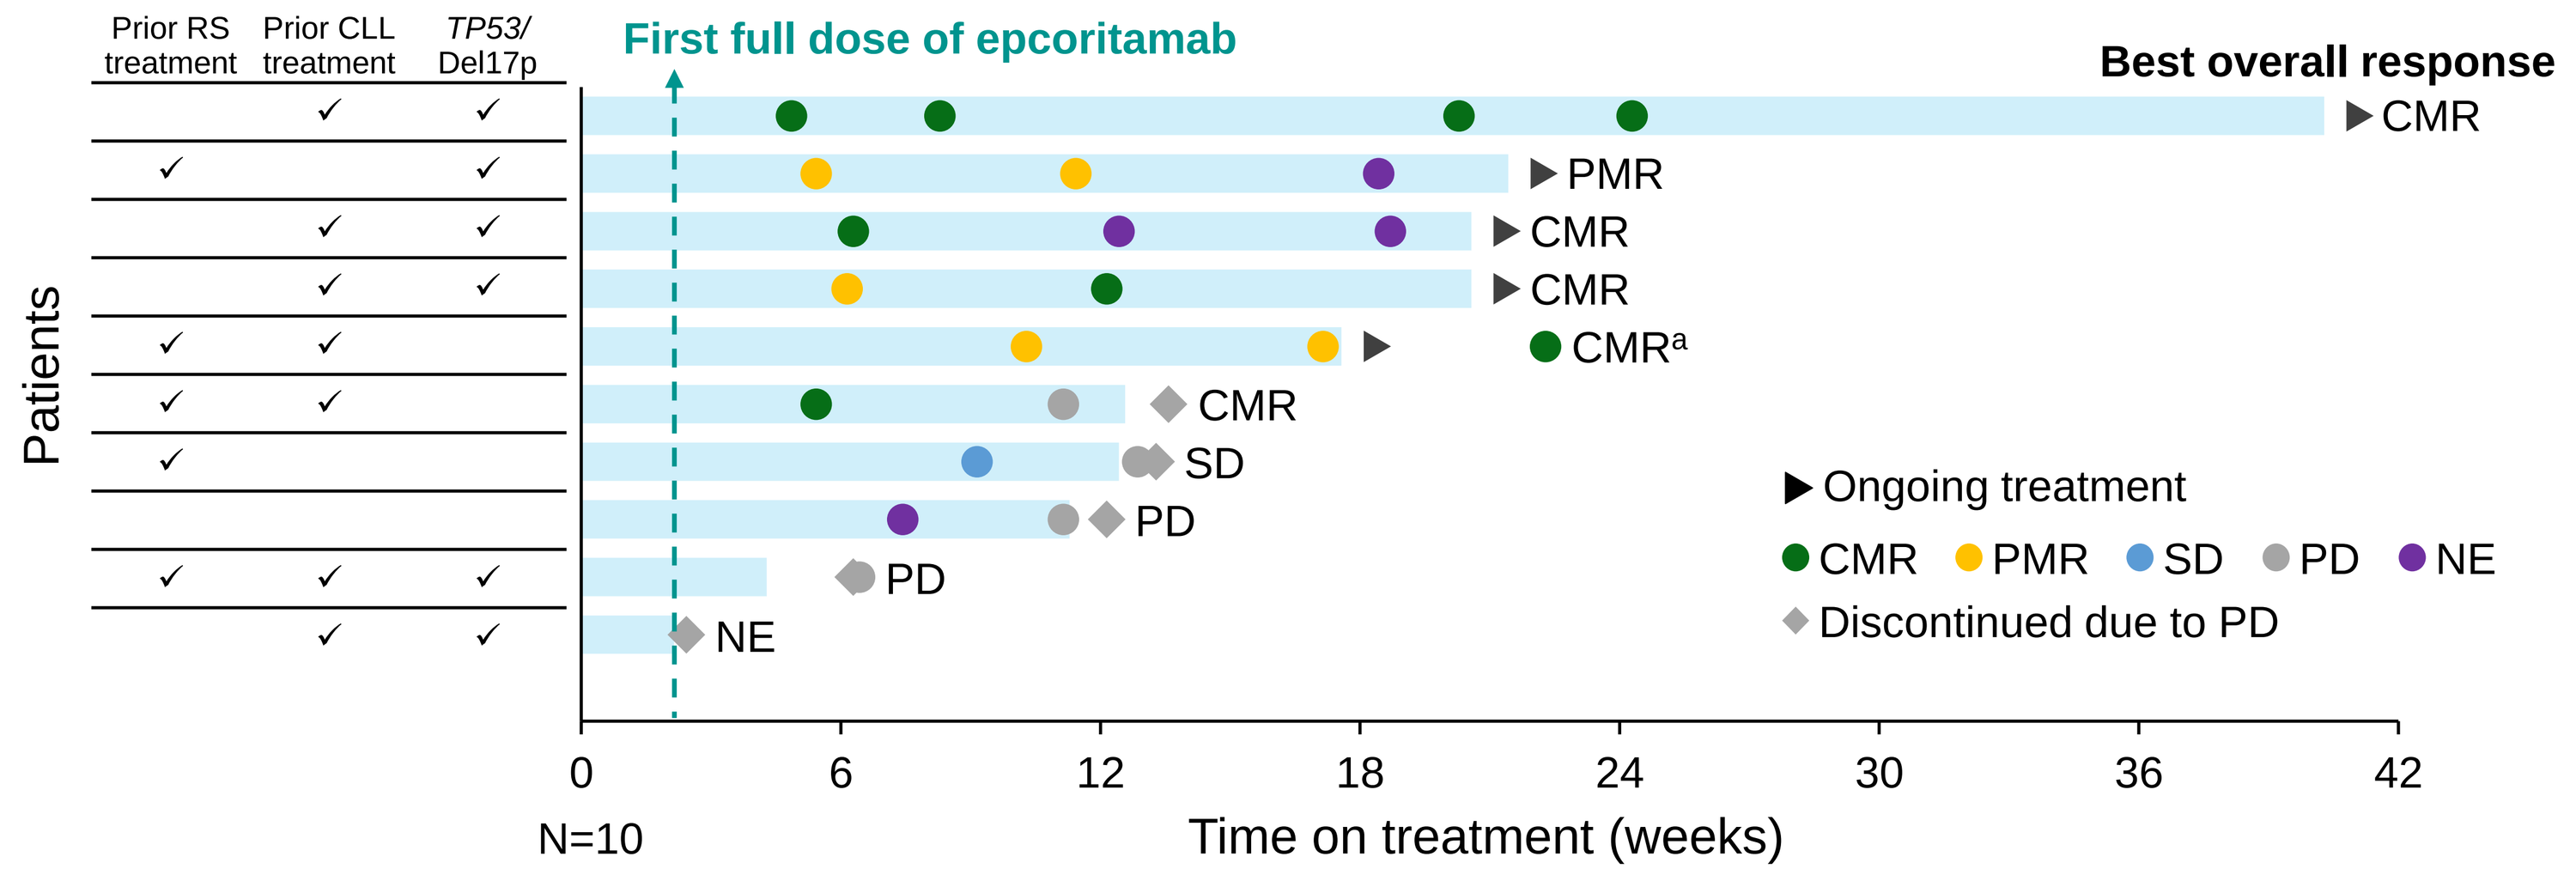
Prior RS treatment
Prior CLL treatment
TP53/ Del17p
First full dose of epcoritamab
Best overall response
Patients
CMR
PMR
CMR
CMR
CMR[a]
CMR
SD
PD
PD
NE
Ongoing treatment
CMR PMR SD PD NE
Discontinued due to PD
N=10
Time on treatment (weeks)

Data cutoff: September 8, 2022. Median follow-up: 4.9 mo (range, 0.6–9.3). Tumor response was evaluated by PET-CT obtained Q6W until wk 24, and then Q24W until disease progression. Per protocol, patients continued to receive scans if they discontinued treatment for reasons other than PD. Check marks indicate patient characteristics. [a]Response was verified as CMR post data cutoff, with updated bone marrow results, while patient was receiving 40 mg of prednisone.

- Median time to response was 1.3 mo (range, 1.1–2.4)
- Median time to complete response was 1.4 mo (range, 1.1–2.8)

EPCORE NHL-1 Clinical Case Study: RS-DLBCL

Patient history

- 76-year-old male
- Diagnosed with SLL in Jul 2019, started on ibrutinib
- Transformed to RS-DLBCL in Oct 2020
- RS-DLBCL treated with 3 cycles of R-CHOP, mixed response

Epcoritamab treatment

- First dose: Feb 2021 (SPD = 105 cm^2)
- CR at week 6, 12, 17, 23, 36, 48, 62, 76 (DS = 1, SPD = 2.8 cm^2)
- Patient has been in sustained CR for over 70 weeks and is still on treatment (last dose C22D1)

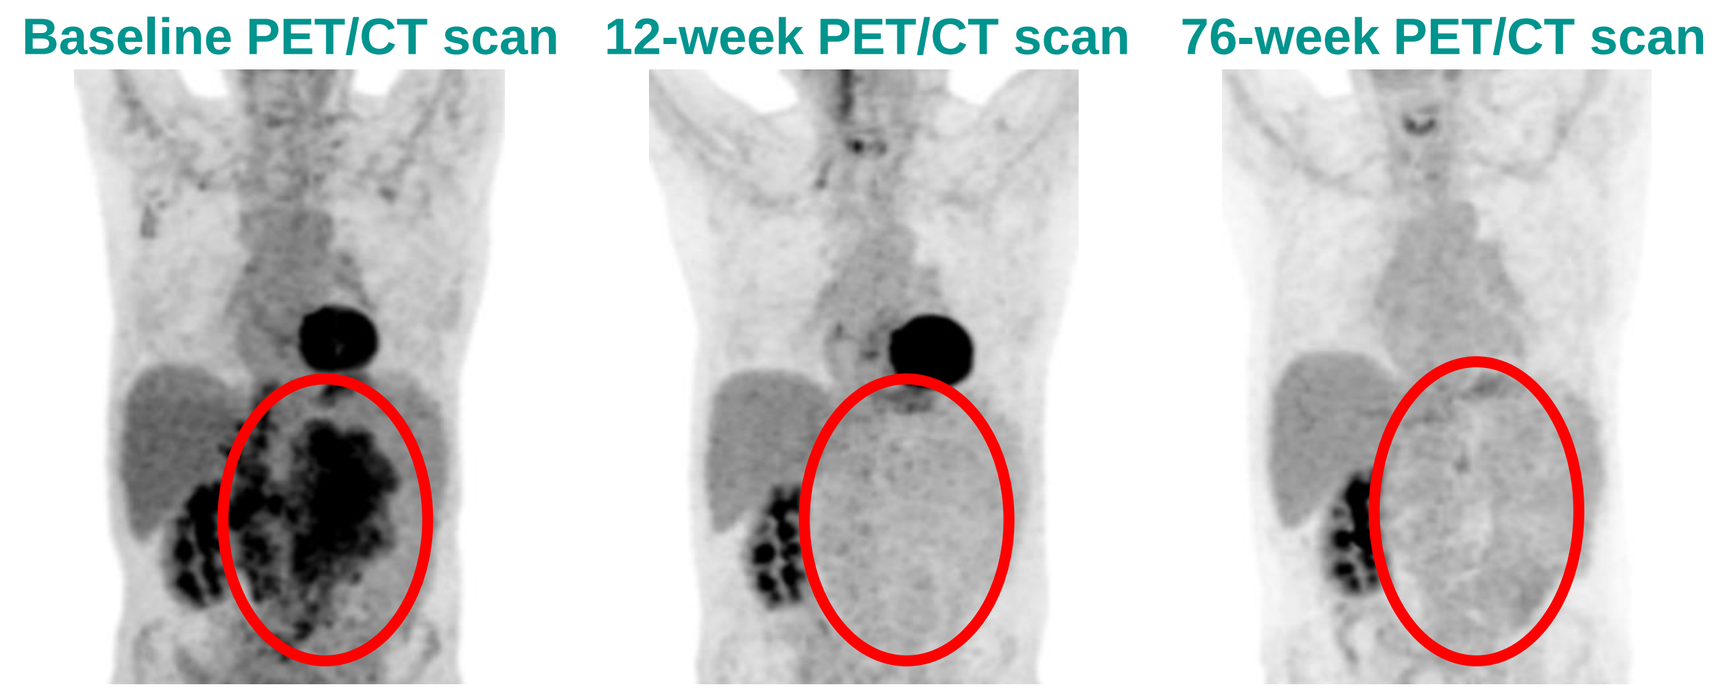
Baseline PET/CT scan
12-week PET/CT scan
76-week PET/CT scan

Mesenteric mass: 11.6 x 7.2 cm — SUV max 6.3 in abdominal/pelvic mass

Mesenteric mass: 0.5 x 0.5 cm — SUV max 2.0 in abdominal/pelvic mass

Mesenteric mass: 0.5 x 0.5 cm — Background uptake only

Courtesy of Dr. Yasmin H. Karimi.

Presented at the 27th Annual International Congress on Hematologic Malignancies®: Focus on Leukemias, Lymphomas, and Myeloma; February 23–26, 2023; Miami, FL